The photodegradation of nifedipine and its cocrystals

Samuel A Stratford*[a], Mark D Eddleston[a], Cheryl Doherty[b], Garry Scrivens[b], William Jones[a]

* - sas82@cam.ac.uk, a – Department of Chemistry, University of Cambridge. b – Pfizer UK, Sandwich.

Abstract

A range of cocrystals of the photosensitive drug nifedipine were prepared and characterised. The rates of solid-state degradation were measured and a correlation was observed between the absorption properties of a cocrystal and its degradation rate. The use of AFM demonstrated the significant changes occurring at the surface during degradation as well as the formation of an amorphous layer before crystallisation of the degradant.

Background & cocrystal structures

Nifedipine (NIF) is a dihydropyridine drug often used for the treatment of hypertension and angina. When exposed to light (both in solution and in the solid-state) it degrades to two major products, a nitropyridine derivative (minor product) and a nitrosopyridine derivative (formed by loss of water, major product) (Fig. 1)[1].

In order to investigate the solid-state degradation of NIF, a range of cocrystals (Fig. 2) and physical mixtures[#] have been prepared and irradiated for a range of time periods up to 24 hours. A High Performance Liquid Chromatography (HPLC) method was developed to quantify the extent of the degradation of NIF.

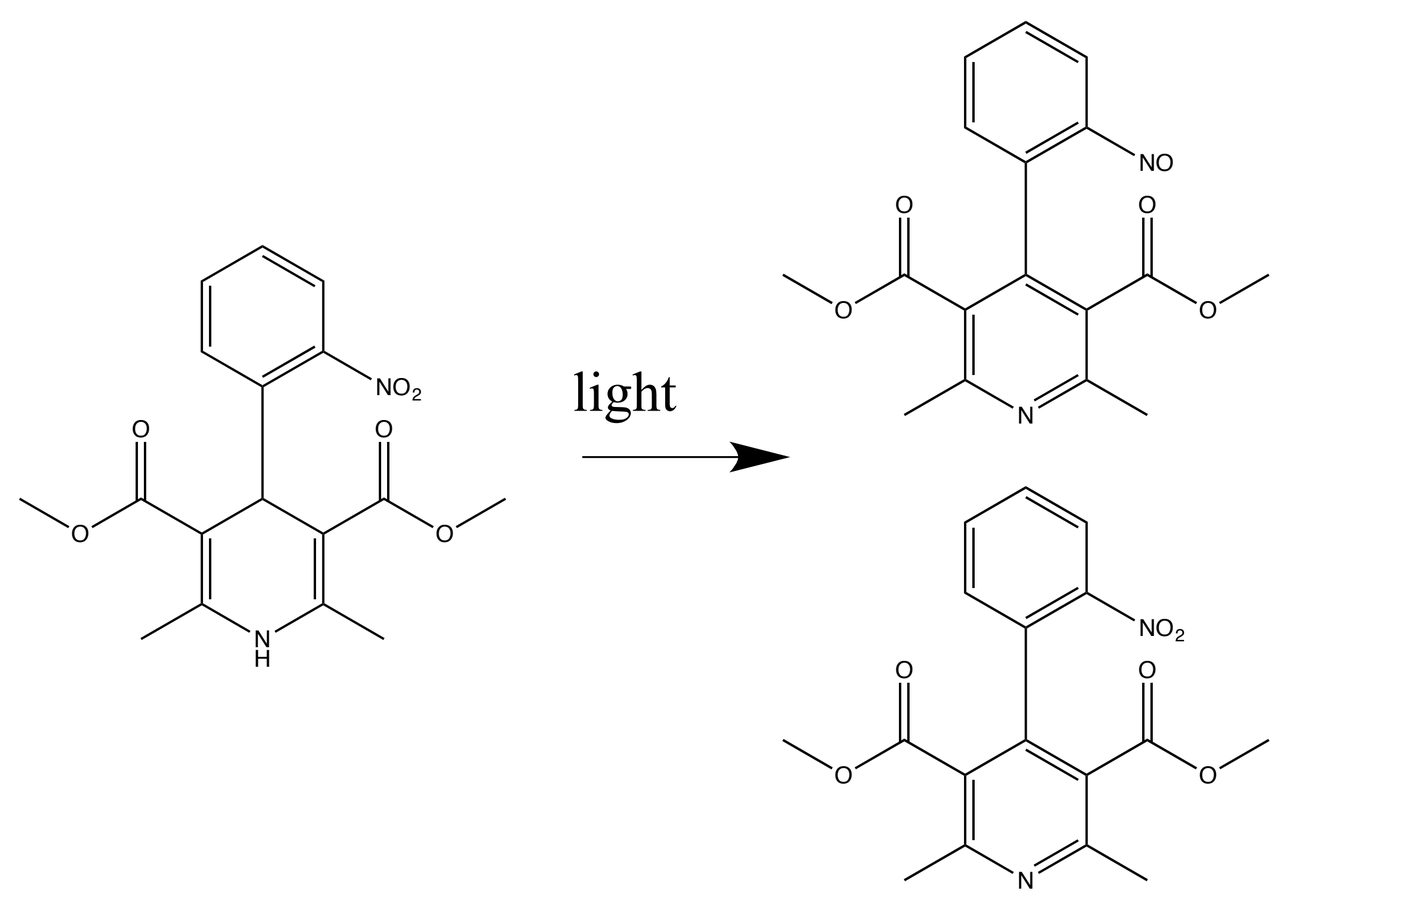
light

Fig. 1 – NIF degrades to nitrosopyridine derivative (major, top) and nitropyridine derivative (minor, bottom)

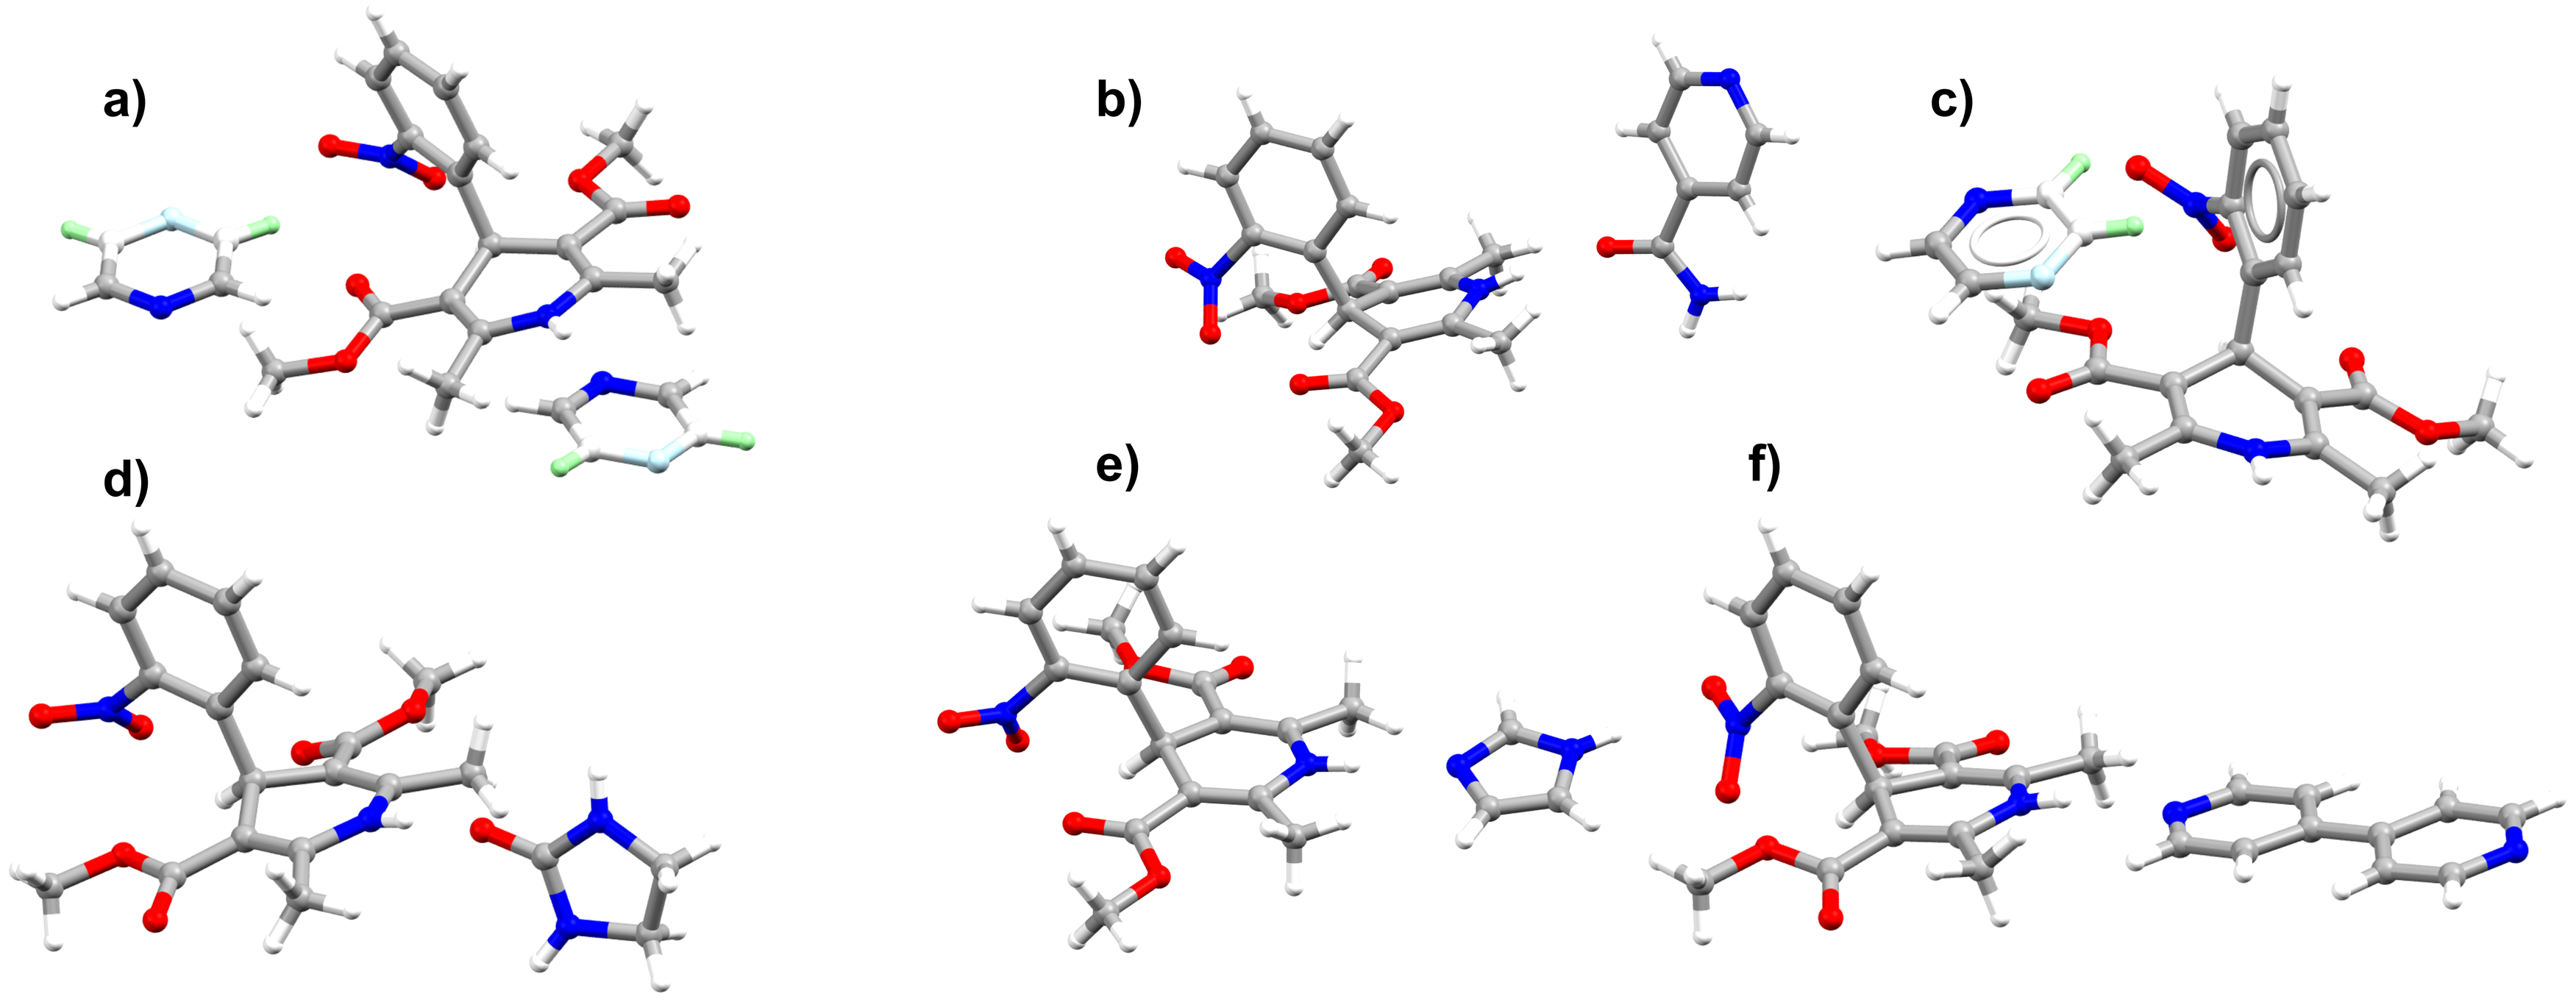
a)
b)
c)
d)
e)
f)

Fig. 2 – cocrystals with NIF – a) NIF·pyrazine (1:1), b) NIF·isonicotinamide[2], c) NIF·pyrazine (2:1, CSD ref: SUZGEA)[3], d) NIF·2-imidazolidone, e) NIF·imidazole, f) NIF·4,4'dipyridyl[2]

- a physical mixture with pyrazine was unable to be prepared as spontaneous partial cocrystallisation occurred

Degradation Results

Figure 3 highlights the behaviours of the different cocrystals upon irradiation when compared to NIF, whilst the extent of degradation for the physical mixtures appears to remain relatively consistent across all coformers. This shows that it is not just the presence of the coformer molecule that affects the degradation but it is the intimate relationship found in a cocrystal that is more important.

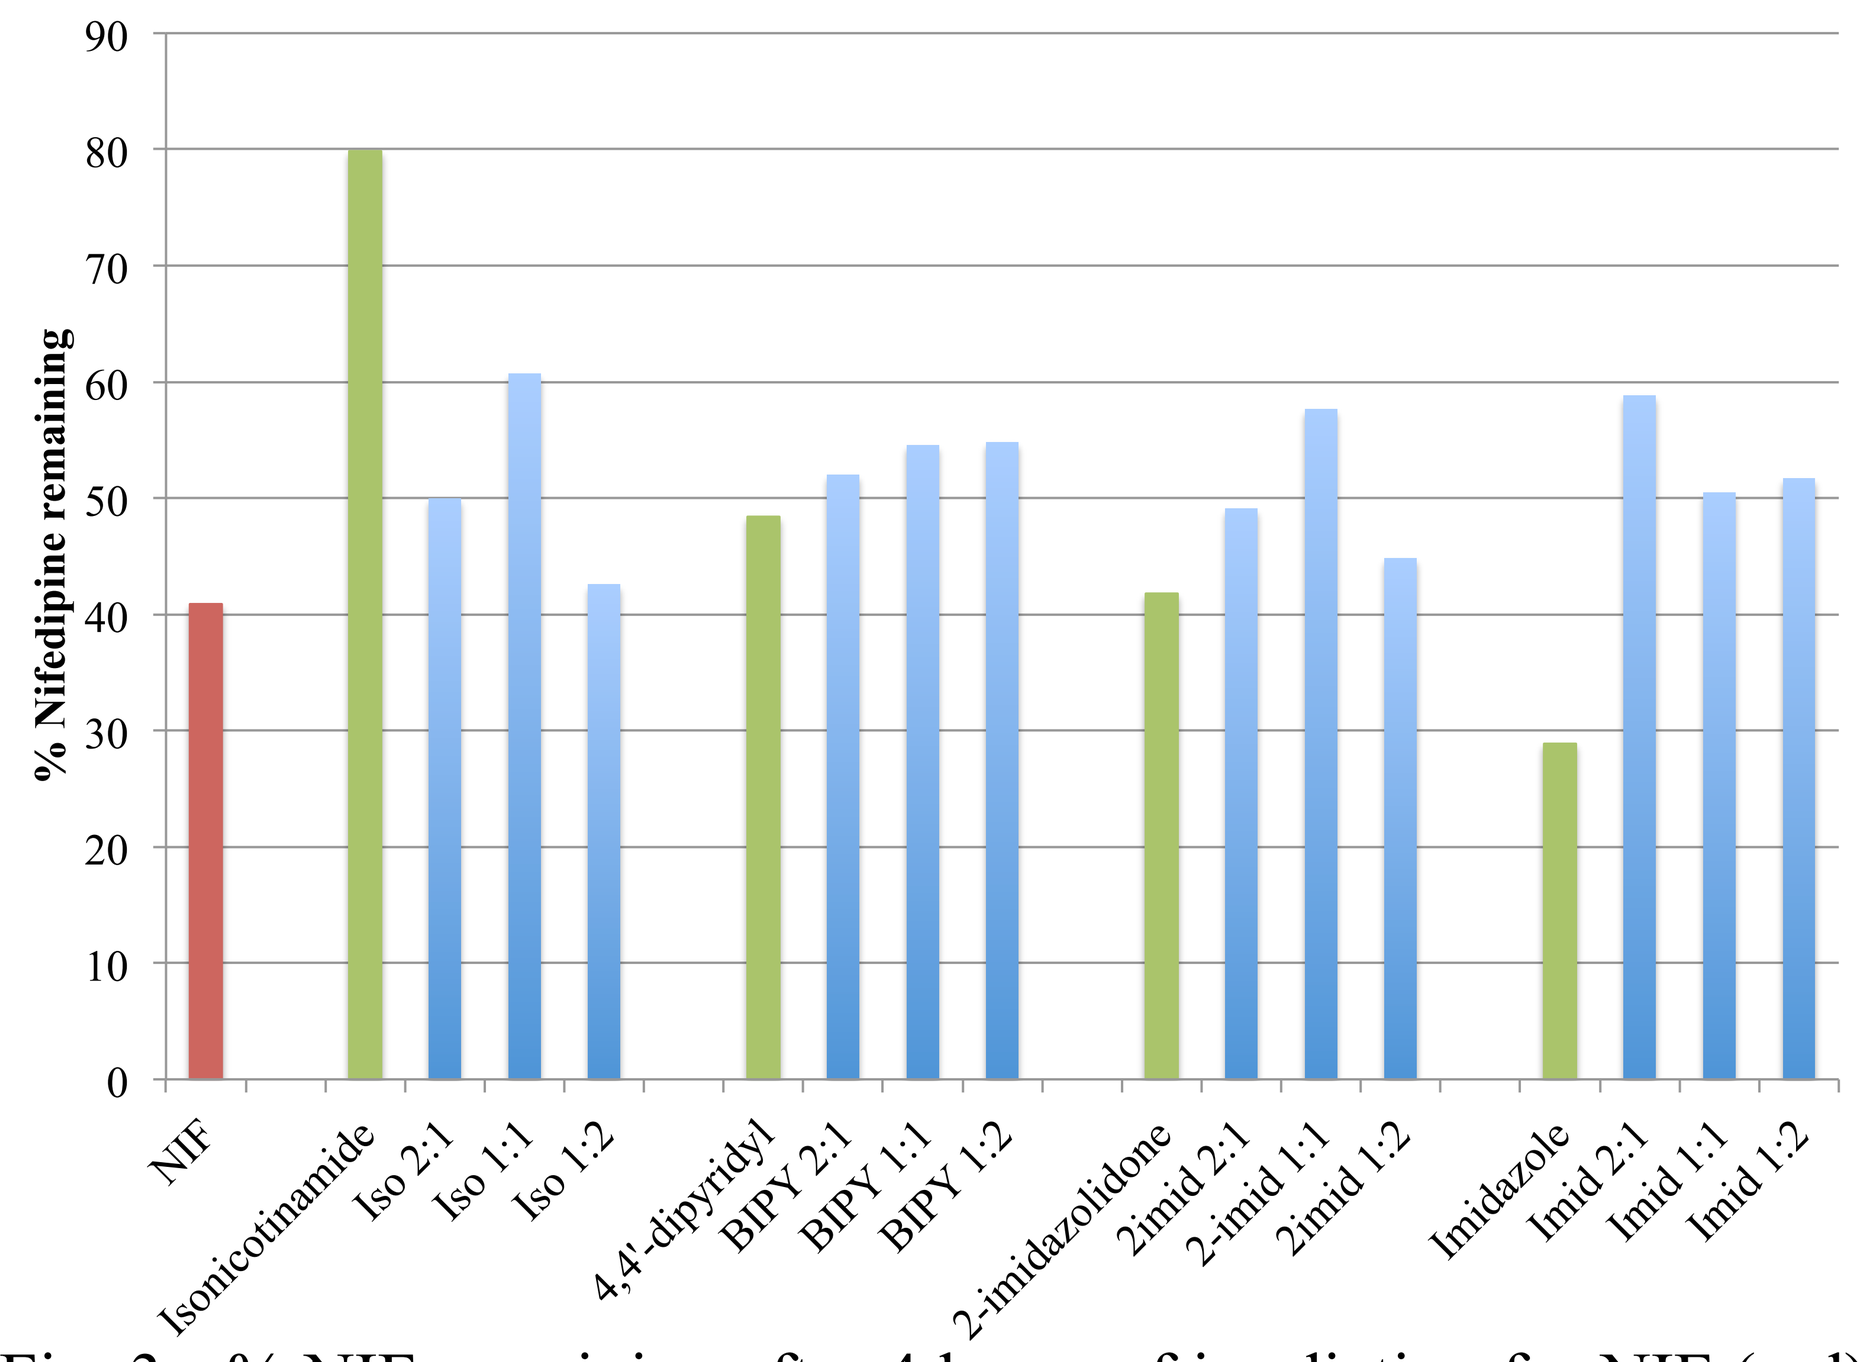
% Nifedipine remaining
NIF
Isonicotinamide
Iso 2:1
Iso 1:1
Iso 1:2
4,4'-dipyridyl
BIPY 2:1
BIPY 1:1
BIPY 1:2
2-imidazolidone
2imid 2:1
2-imid 1:1
2imid 1:2
Imidazole
Imid 2:1
Imid 1:1
Imid 1:2

Fig. 3 - % NIF remaining after 4 hours of irradiation for NIF (red), cocrystals (green) and physical mixtures in different ratios (blue)

The curves shown in Figure 4 illustrate the degree of degradation for NIF and its cocrystals over a range of time periods up to 24 hours. There are clearly some cocrystals that degrade slower than NIF (NIF·isonicotinamide and 2:1 NIF·pyrazine) as well as those that degrade at around the same rate or even quicker than NIF (NIF·imidazole).

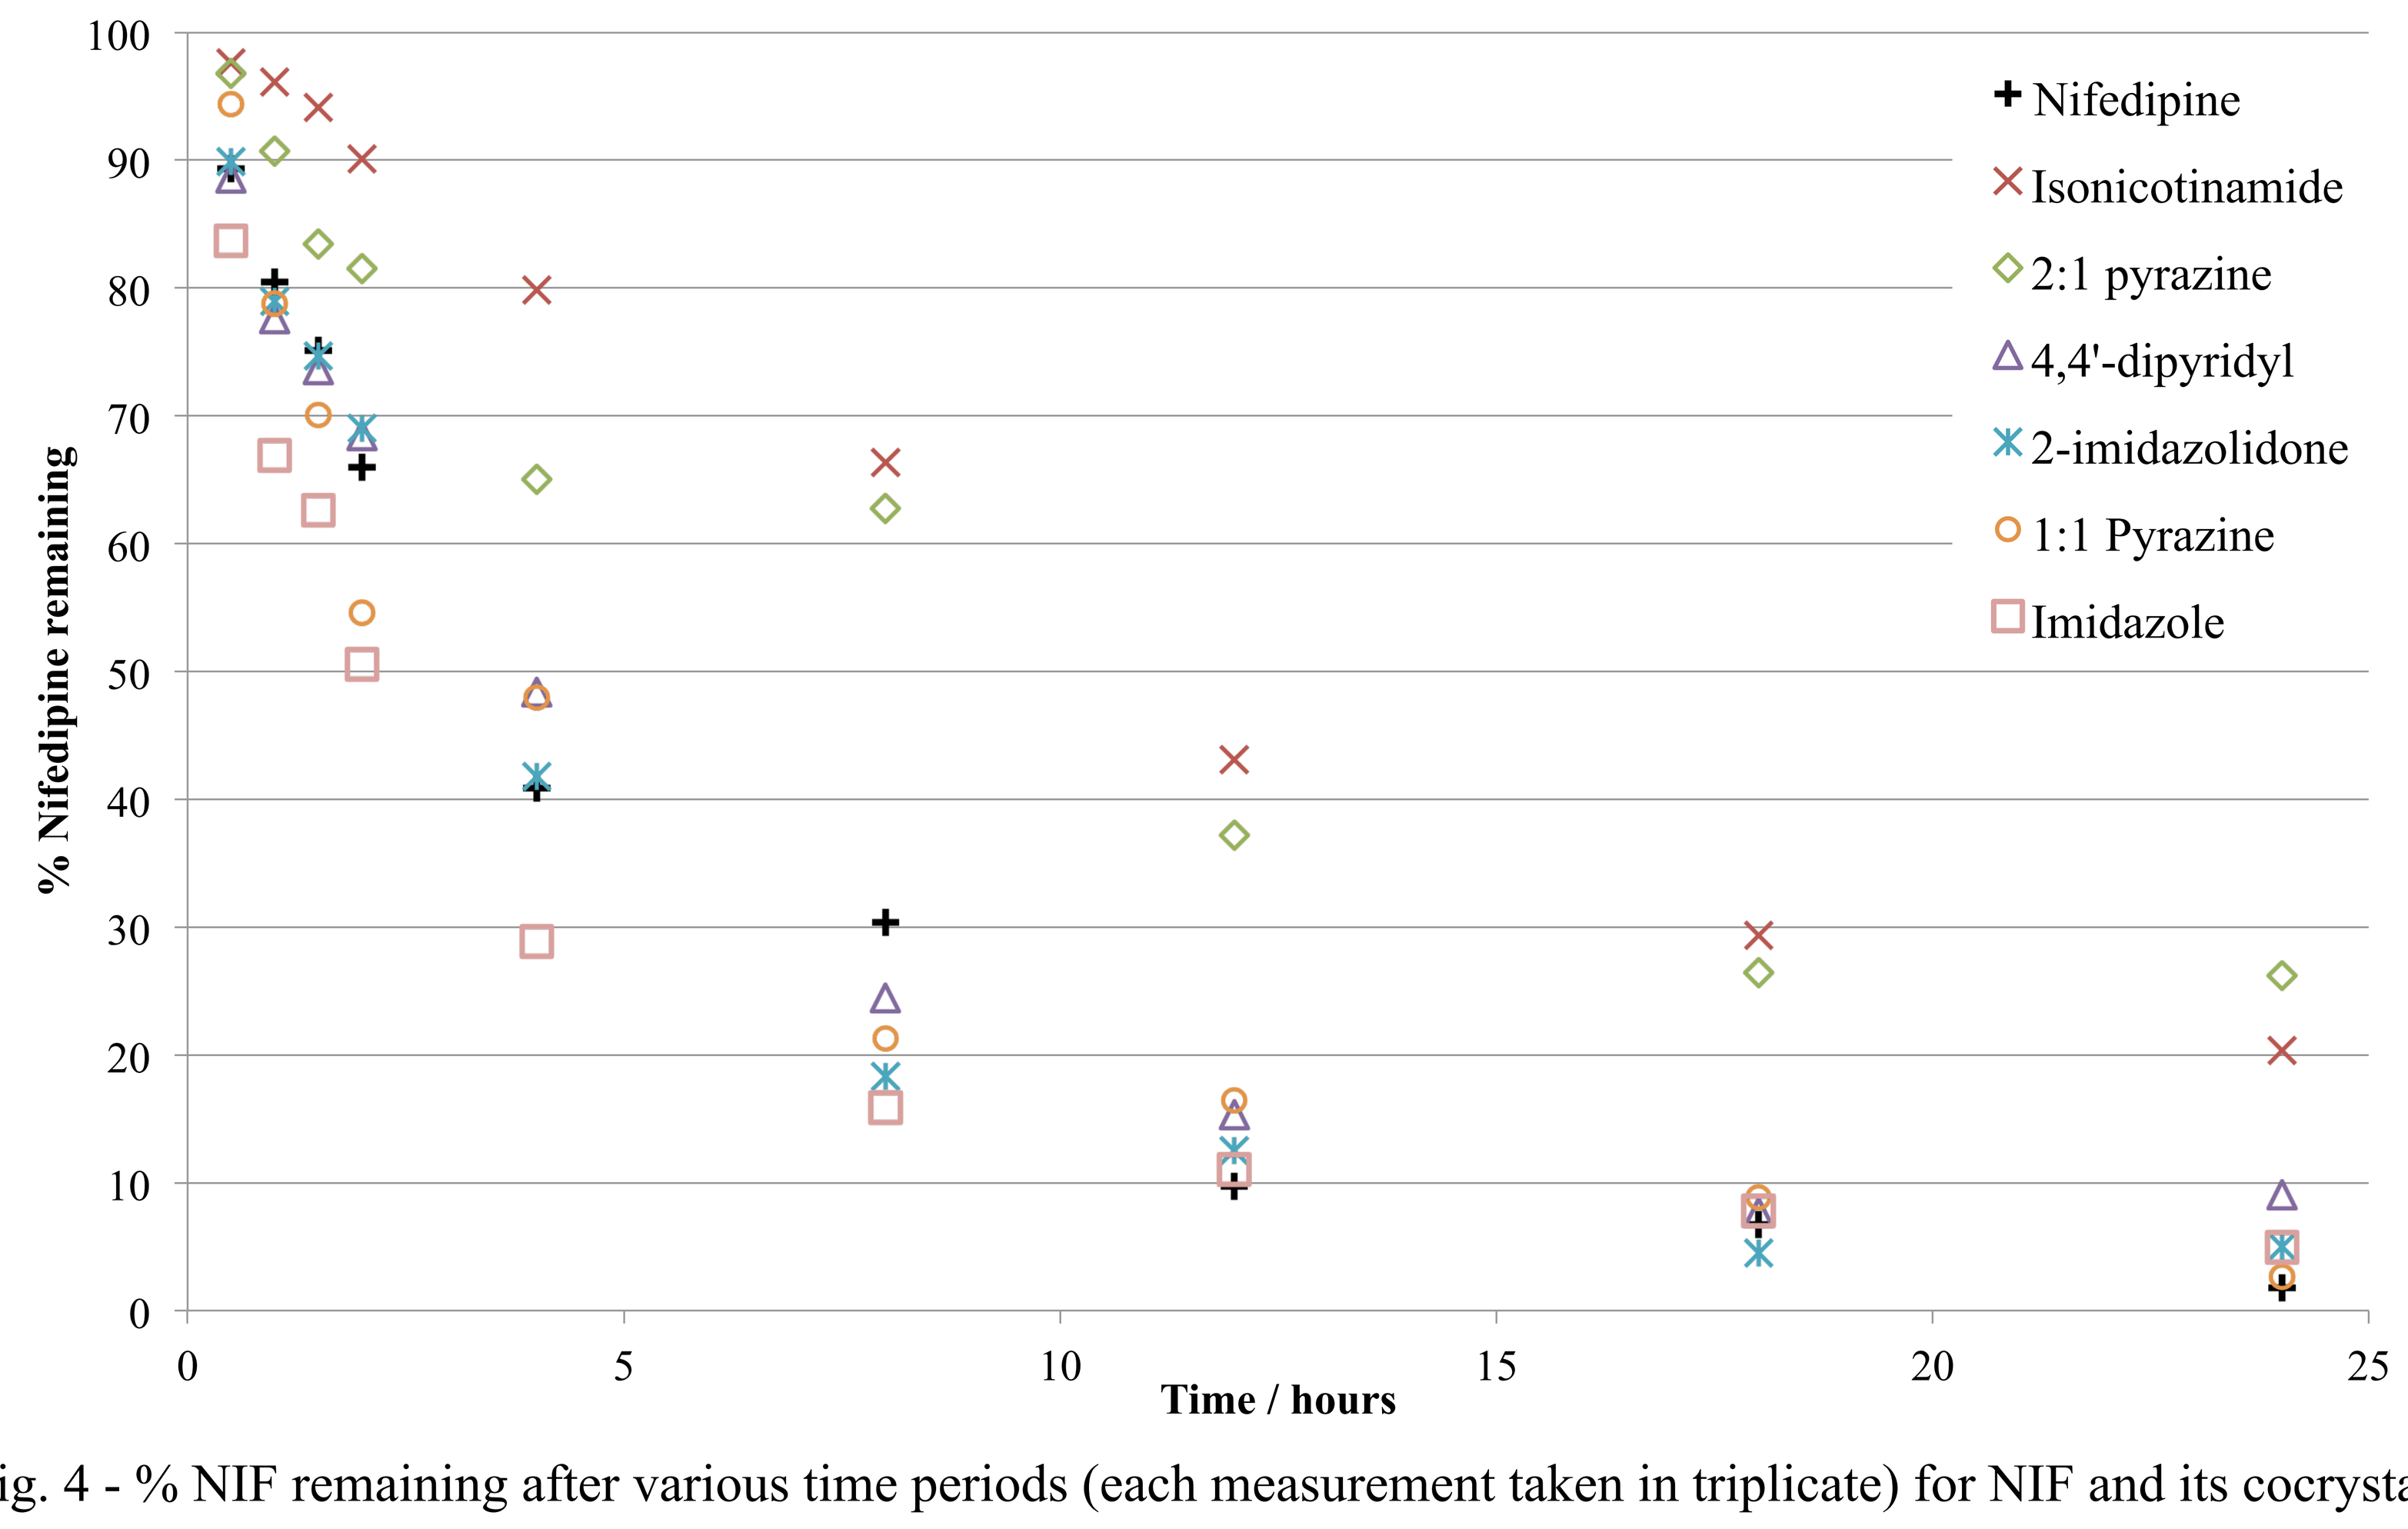
% Nifedipine remaining
Time / hours
Nifedipine
Isonicotinamide
2:1 pyrazine
4,4'-dipyridyl
2-imidazolidone
1:1 Pyrazine
Imidazole

Fig. 4 - % NIF remaining after various time periods (each measurement taken in triplicate) for NIF and its cocrystals

Solid-state UV/vis spectroscopy

As the curves in figure 5a show, there is very little difference in the absorption properties of NIF when compared with its physical mixtures with coformers. In contrast, figure 5b indicates that there is a large difference in the absorption properties of the cocrystals. This is further evidence that it is not just the presence of the coformer molecule that is important but that by influencing the crystal packing of NIF, we can fine-tune its absorption properties.

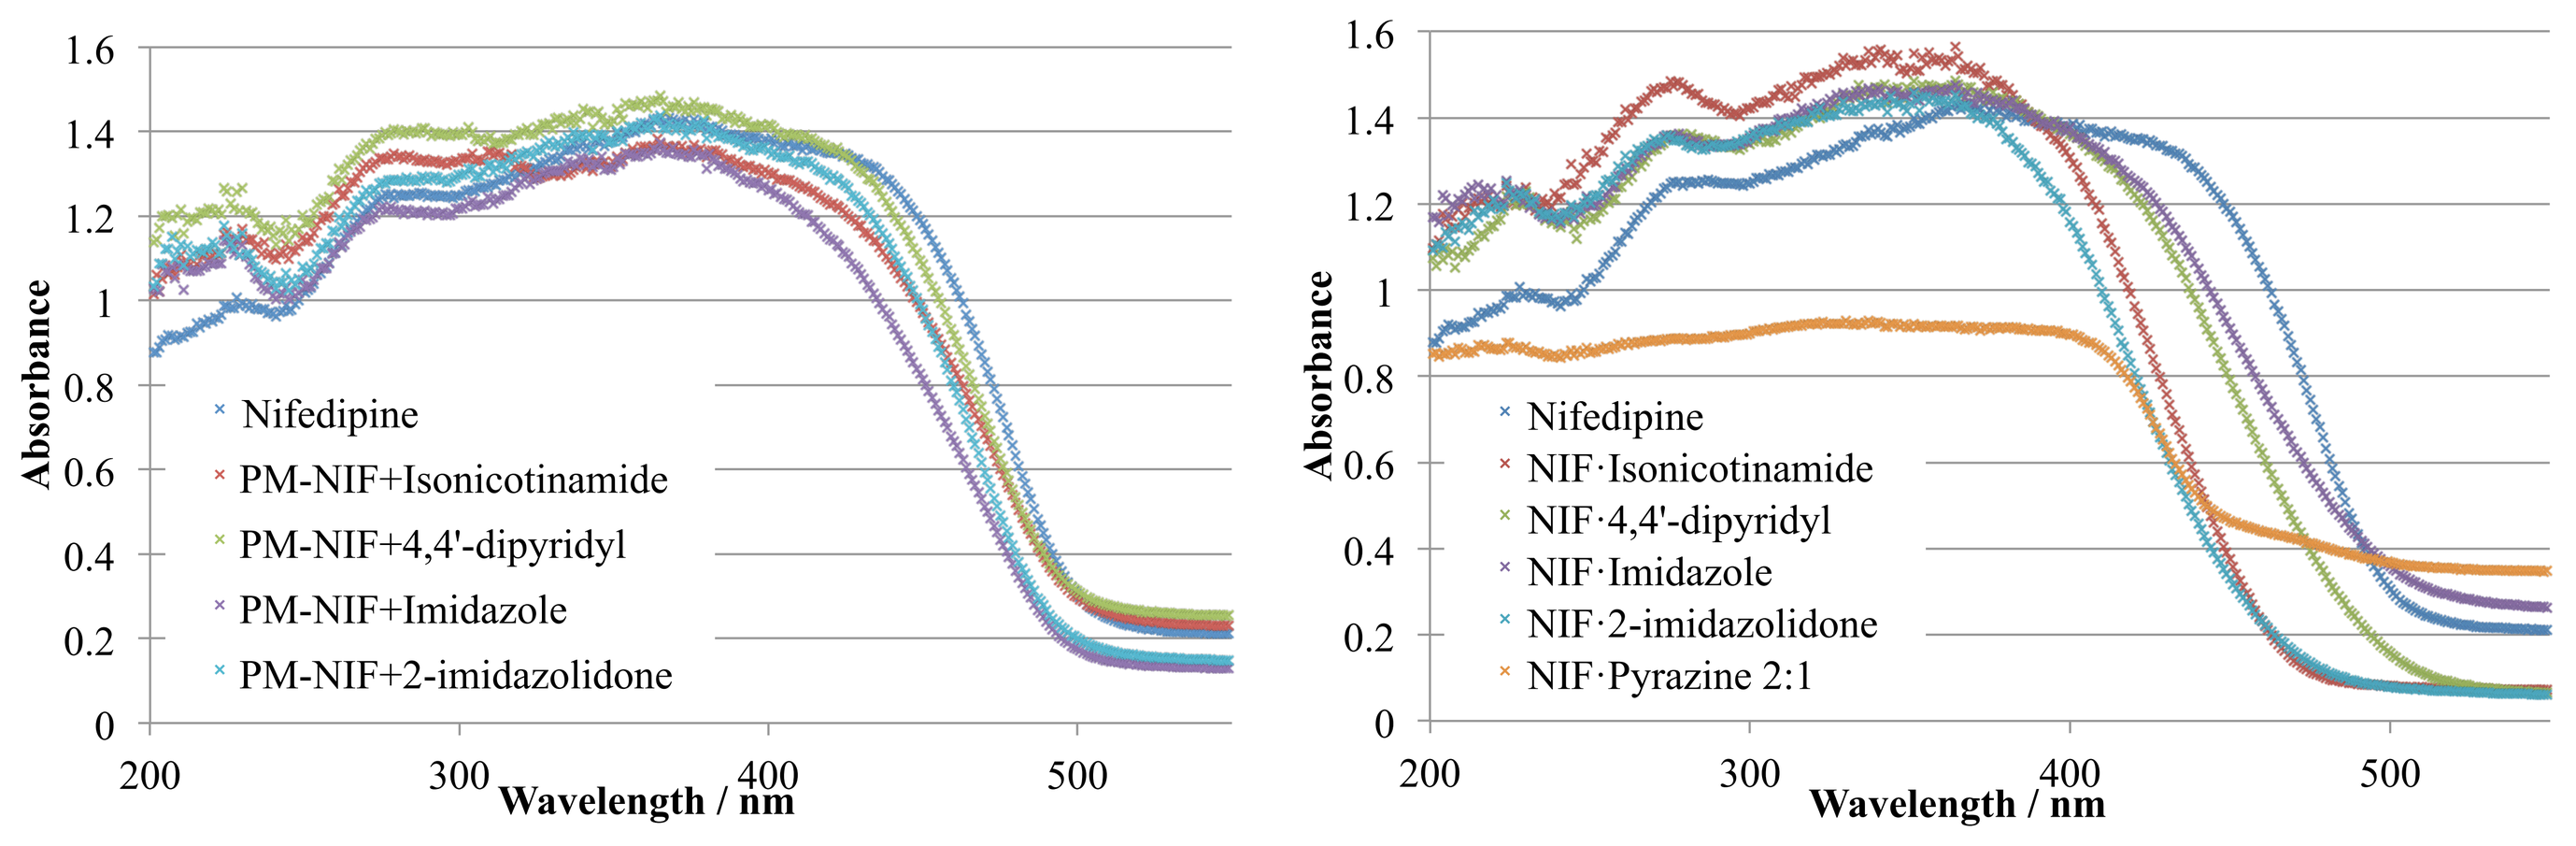
Absorbance
Wavelength / nm
Nifedipine
PM-NIF+Isonicotinamide
PM-NIF+4,4'-dipyridyl
PM-NIF+Imidazole
PM-NIF+2-imidazolidone
Nifedipine
NIF·Isonicotinamide
NIF·4,4'-dipyridyl
NIF·Imidazole
NIF·2-imidazolidone
NIF·Pyrazine 2:1

Fig. 5 – a) Absorption curves for NIF and 1:1 physical mixtures of NIF with coformers, all curves are similar. b) Absorption curves for NIF and its cocrystals, demonstrating the large differences between cocrystals

By plotting a Tauc plot[4] a correlation is observed between the band gap of the material and its rate constant for degradation, the larger the band gap, the slower the degradation (band gap calculated by extrapolating straight line to x-axis).

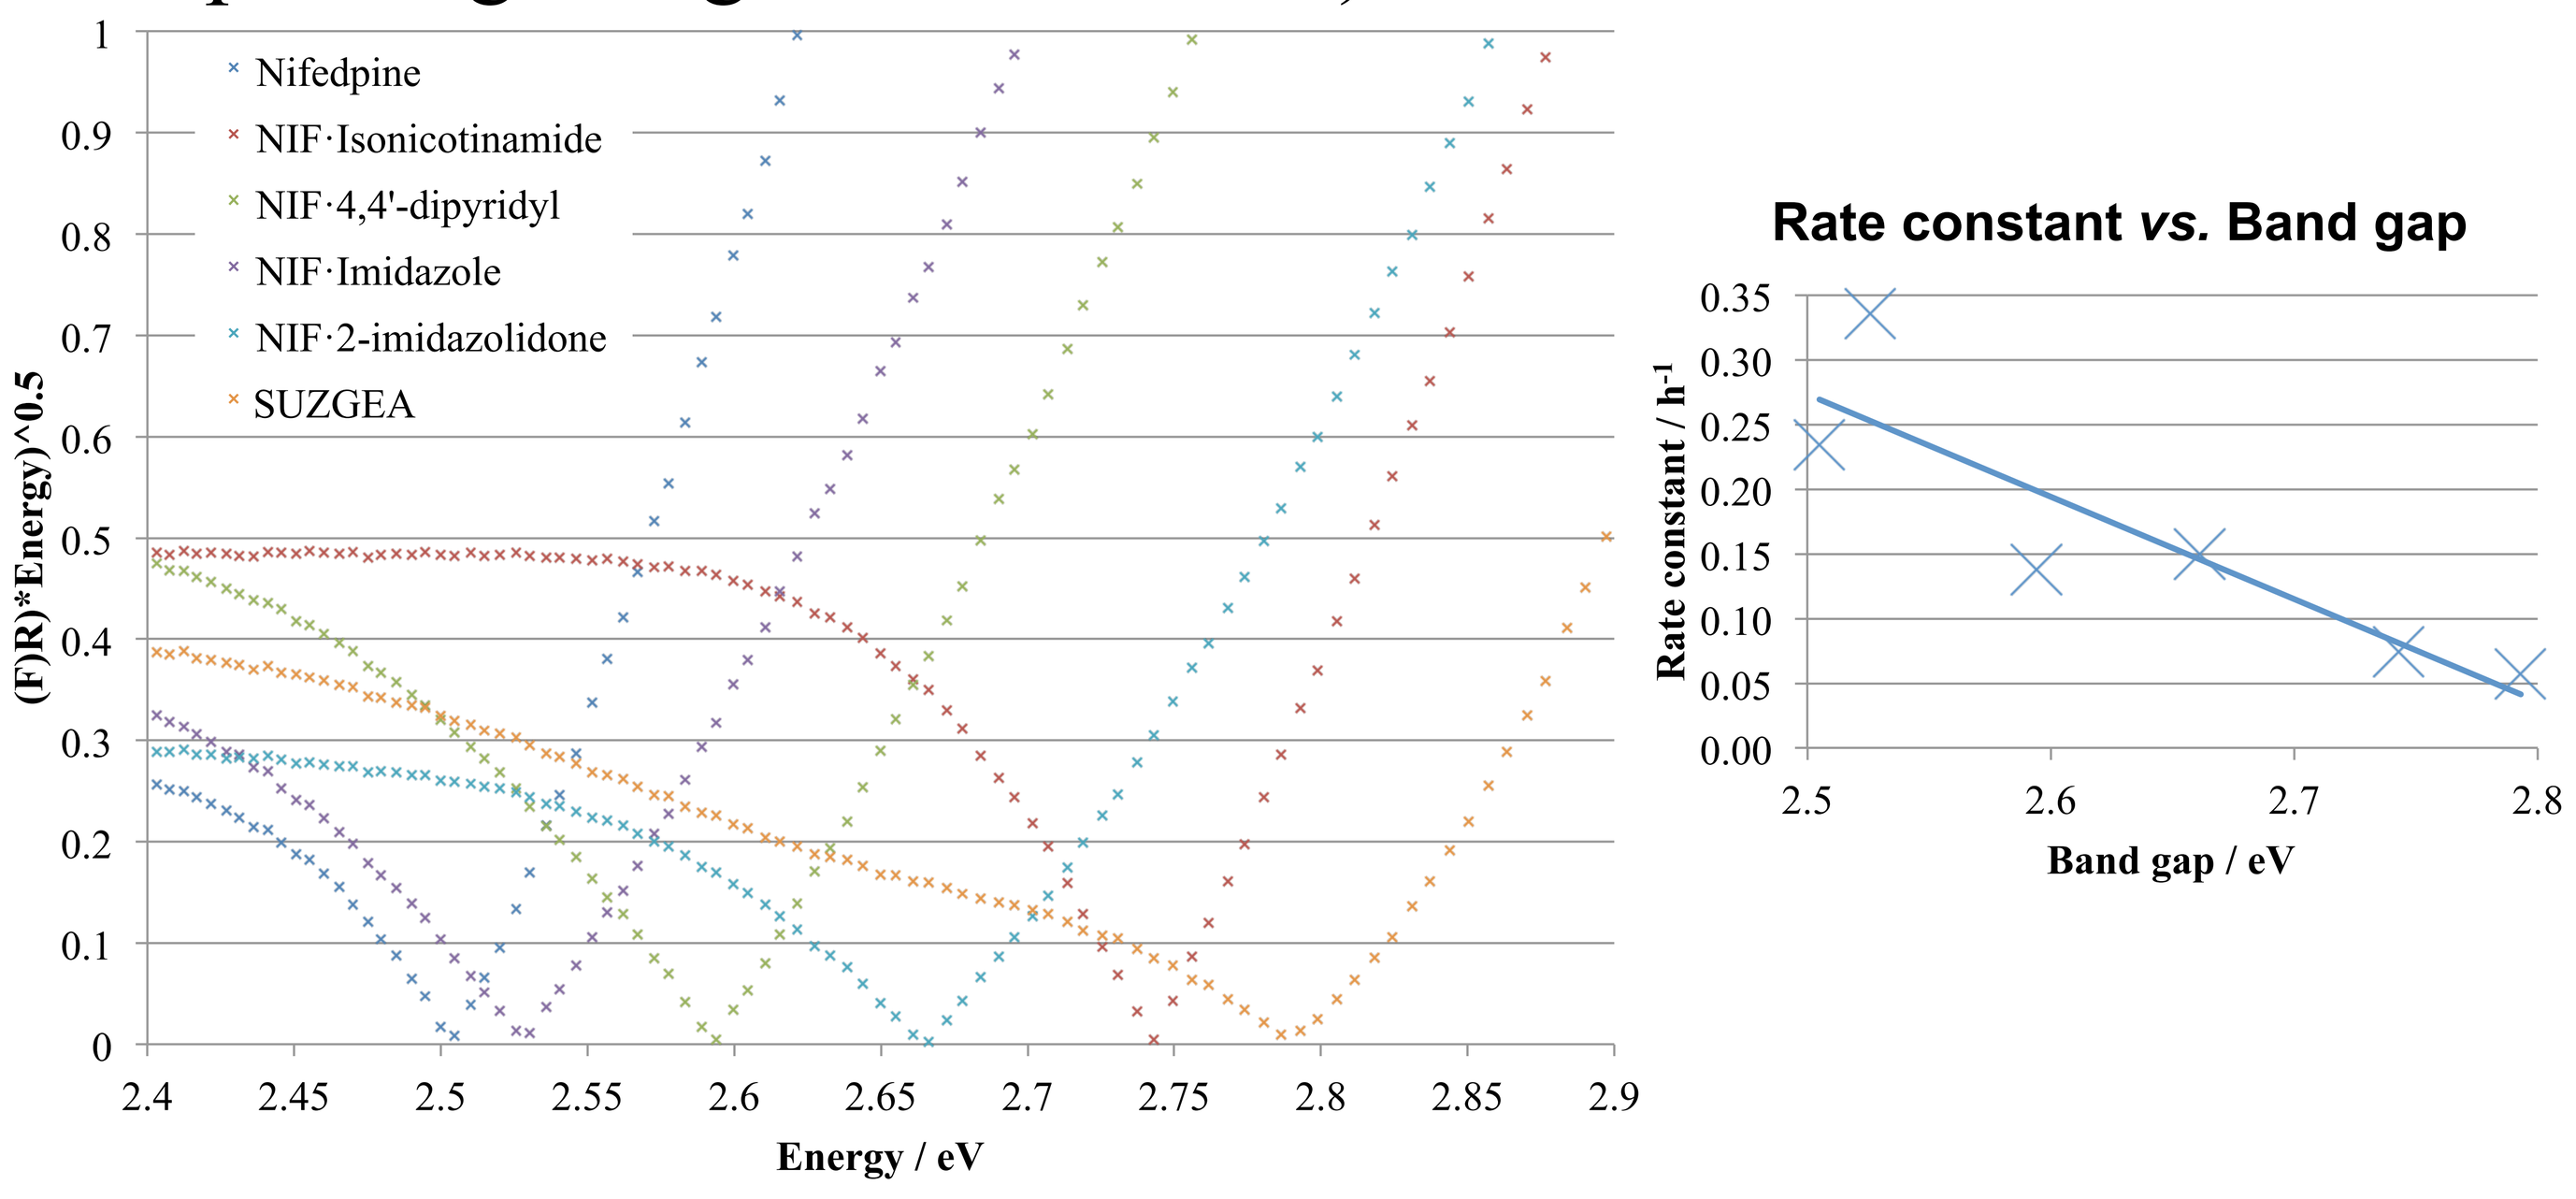
(F)R)*Energy)^0.5
Energy / eV
Nifedpine
NIF·Isonicotinamide
NIF·4,4'-dipyridyl
NIF·Imidazole
NIF·2-imidazolidone
SUZGEA
Rate constant vs. Band gap
Rate constant / h-1
Band gap / eV

Fig. 6 – a) Tauc plot for NIF and its cocrystals. The extrapolation of the straight line to the x-axis corresponds to the band gap. b) The rate constant for degradation plotted against the band gap for NIF and its cocrystals indicating that the larger the band gap, the slower the rate of degradation.

AFM

As shown in figure 7, the surface of nifedipine itself consists of molecular steps (1-2 nm in size). When exposed to light, this surface changes drastically. Initially, an amorphous layer is formed across the surface before crystallisation of the degradant occurs at multiple different nucleation points. The surface is now highly disordered with step sizes ranging from 50-100 nm.

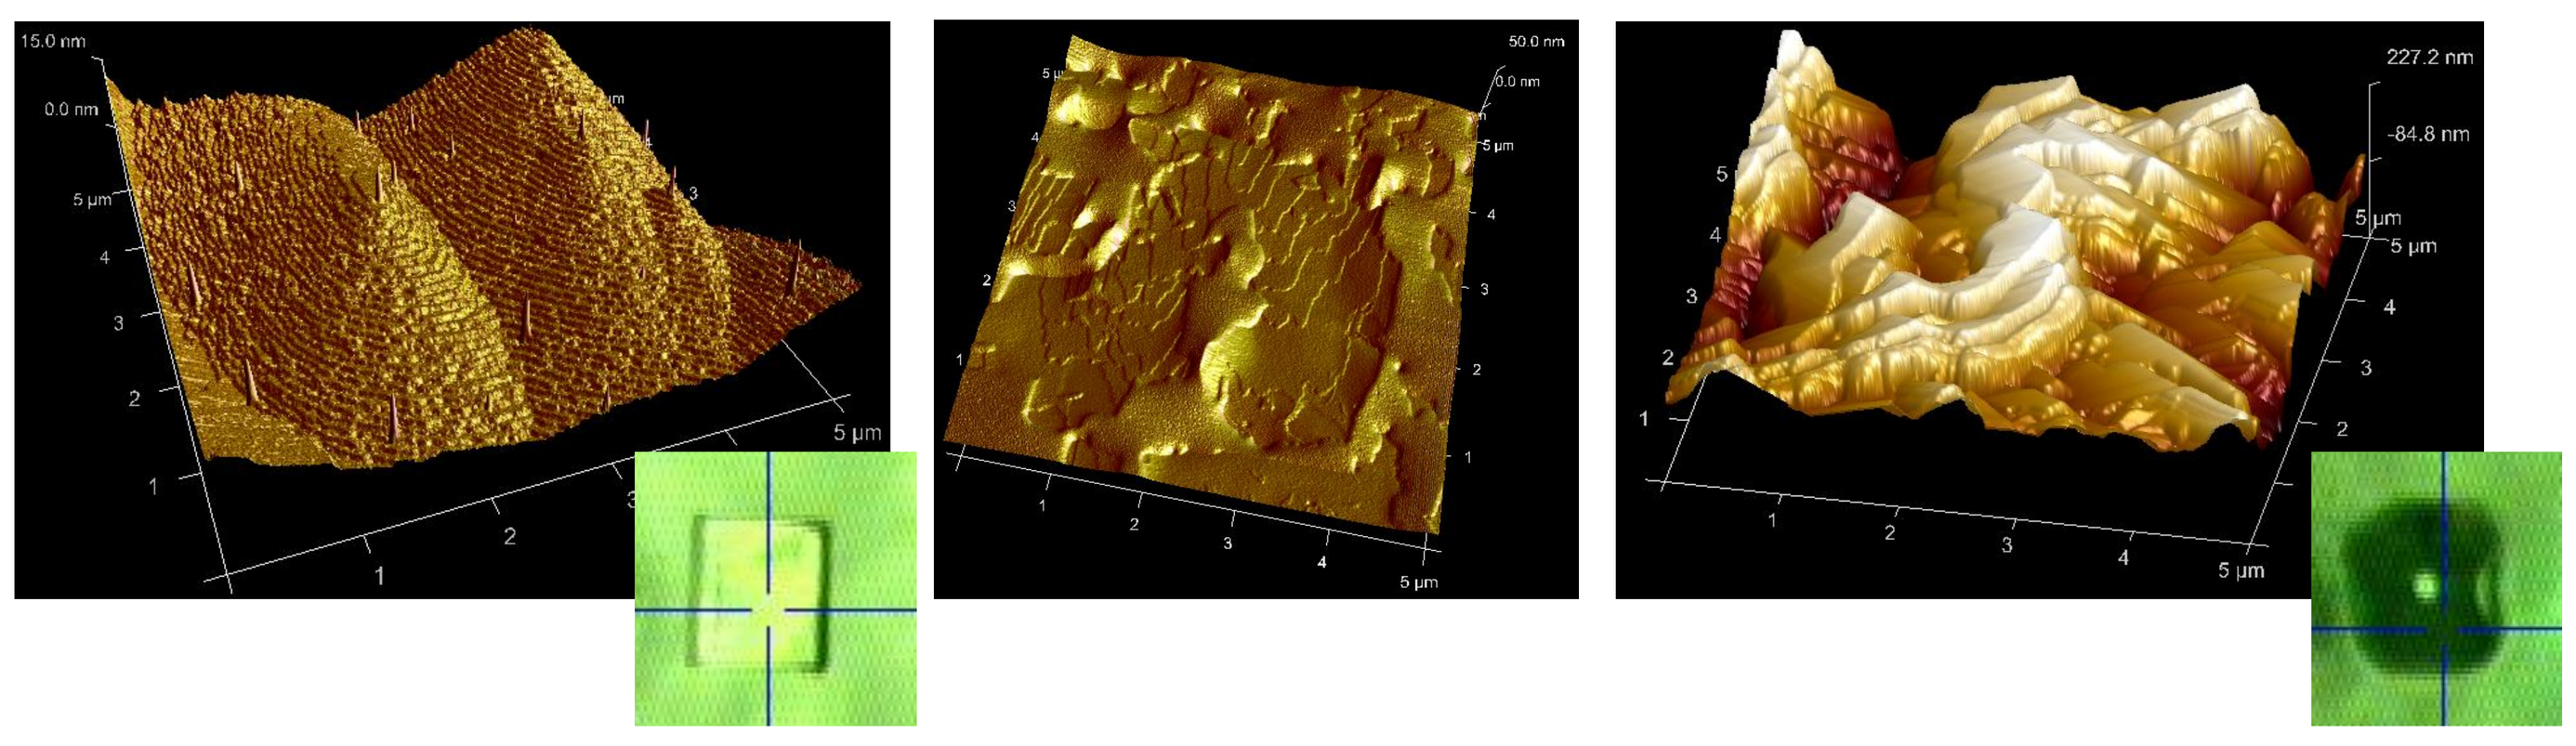

Fig. 7 – AFM height images: a) NIF pre-degradation showing molecular steps. b) NIF during degradation, (flat parts are amorphous). c) NIF post-degradation, surface is highly disordered indicating multiple nucleation points. (inset- optical microscope images of NIF pre- and post-irradiation)

Conclusions

The optical absorption properties of a photolabile drug molecule have been fine-tuned by crystal engineering methods. These properties have been linked to the rate constants for its degradation with those solid forms that have a larger band gap also having a slower rate of degradation.

AFM has demonstrated that, upon irradiation, an amorphous layer is initially formed at the surface of the drug before crystallisation of the degradant occurs.

References & Acknowledgements

1 – Caira, M. *et al.* J. Pharm. Sci. **2003**, 92, 2519-33

2 – Morales, L. A. Crystal Engineering of Binary Compounds Containing Pharmaceutical Molecules, University of South Florida, **2003**.

3 – Schultheiss, N. *et al.* Acta Cryst. E, **2010**, 66, o2297-8

4 – Tauc, J. Materials Research Bulletin, **1968**, 3, 37-46

SAS thanks EPSRC and Pfizer for a studentship, MDE thanks Pfizer for funding.

EPSRC Engineering and Physical Sciences Research Council · Pfizer · UNIVERSITY OF CAMBRIDGE